Revolutionary Variable Pathlength Spectroscopy for AAV Titer Determination to Accelerate Processes Development

Yan Chen[1], JiaBei Zhang[1], Ying Hou[1], Nitin Garg[1], Harald Ehlen[2], Joe Ferraiolo[2], Gicell Schaenzler[2], Nigel Herbert[2], Paul Mania[2]
[1]Gene Therapy Process Development, Tech Ops, PTC Therapeutics, Inc. [2]Repligen Corporation.

REPLIGEN

Abstract

Recombinant adeno-associated virus (rAAV) vectors have emerged as a promising platform for gene therapy. As the demand for rAAV-based gene therapies grows, reliable and efficient methods for rAAV vector titer quantification become increasingly important, especially in process development and manufacturing. We developed a novel method for measuring AAV titer using Variable Pathlength Technology (VPT). Testing of various downstream in-process samples showed a remarkable linear correlation between vector titer values obtained with the CTech™ SoloVPE® System and technologies predominantly established in the industry at present. The method demonstrated excellent linearity, repeatability, and intermediate precision, with a dynamic testing range from 10^{11} vg/ml up to 10^{15} vg/ml for vector titer measurement. Here, we discuss the applications of VPT in downstream process development and its ability to provide real-time feedback to accelerate the process.

Background

Vector and capsid titers and full/empty ratio are Critical Quality Attributes (CQAs) of rAAVs and are closely tracked during downstream process development. Current industry-accepted rAAV titer assays include qPCR, ddPCR, Capsid ELISA, transmission electron microscopy (TEM), analytical ultracentrifugation (AUC), and some high-performance liquid chromatography (HPLC) methods. These methods have their own advantages; however, assay turnaround can be time consuming. Variable Pathlength Technology is a UV technology that allows for measuring the optical density (OD) of samples by analyzing changes in absorbance over multiple pathlengths of interest, which differs from traditional UV-Vis spectroscopy. Although VPT, using the SoloVPE System, has been widely adopted as a titer measurement method in various biopharmaceutical industries for concentration measurement, its use in gene therapy has been limited.

Methods

Variable pathlength spectroscopy differs from traditional fixed pathlength spectrophotometers in that it uses multiple pathlengths to calculate the optical density (OD) value. It does not rely on absolute absorbance measurements and can provide results quickly without sample dilution.

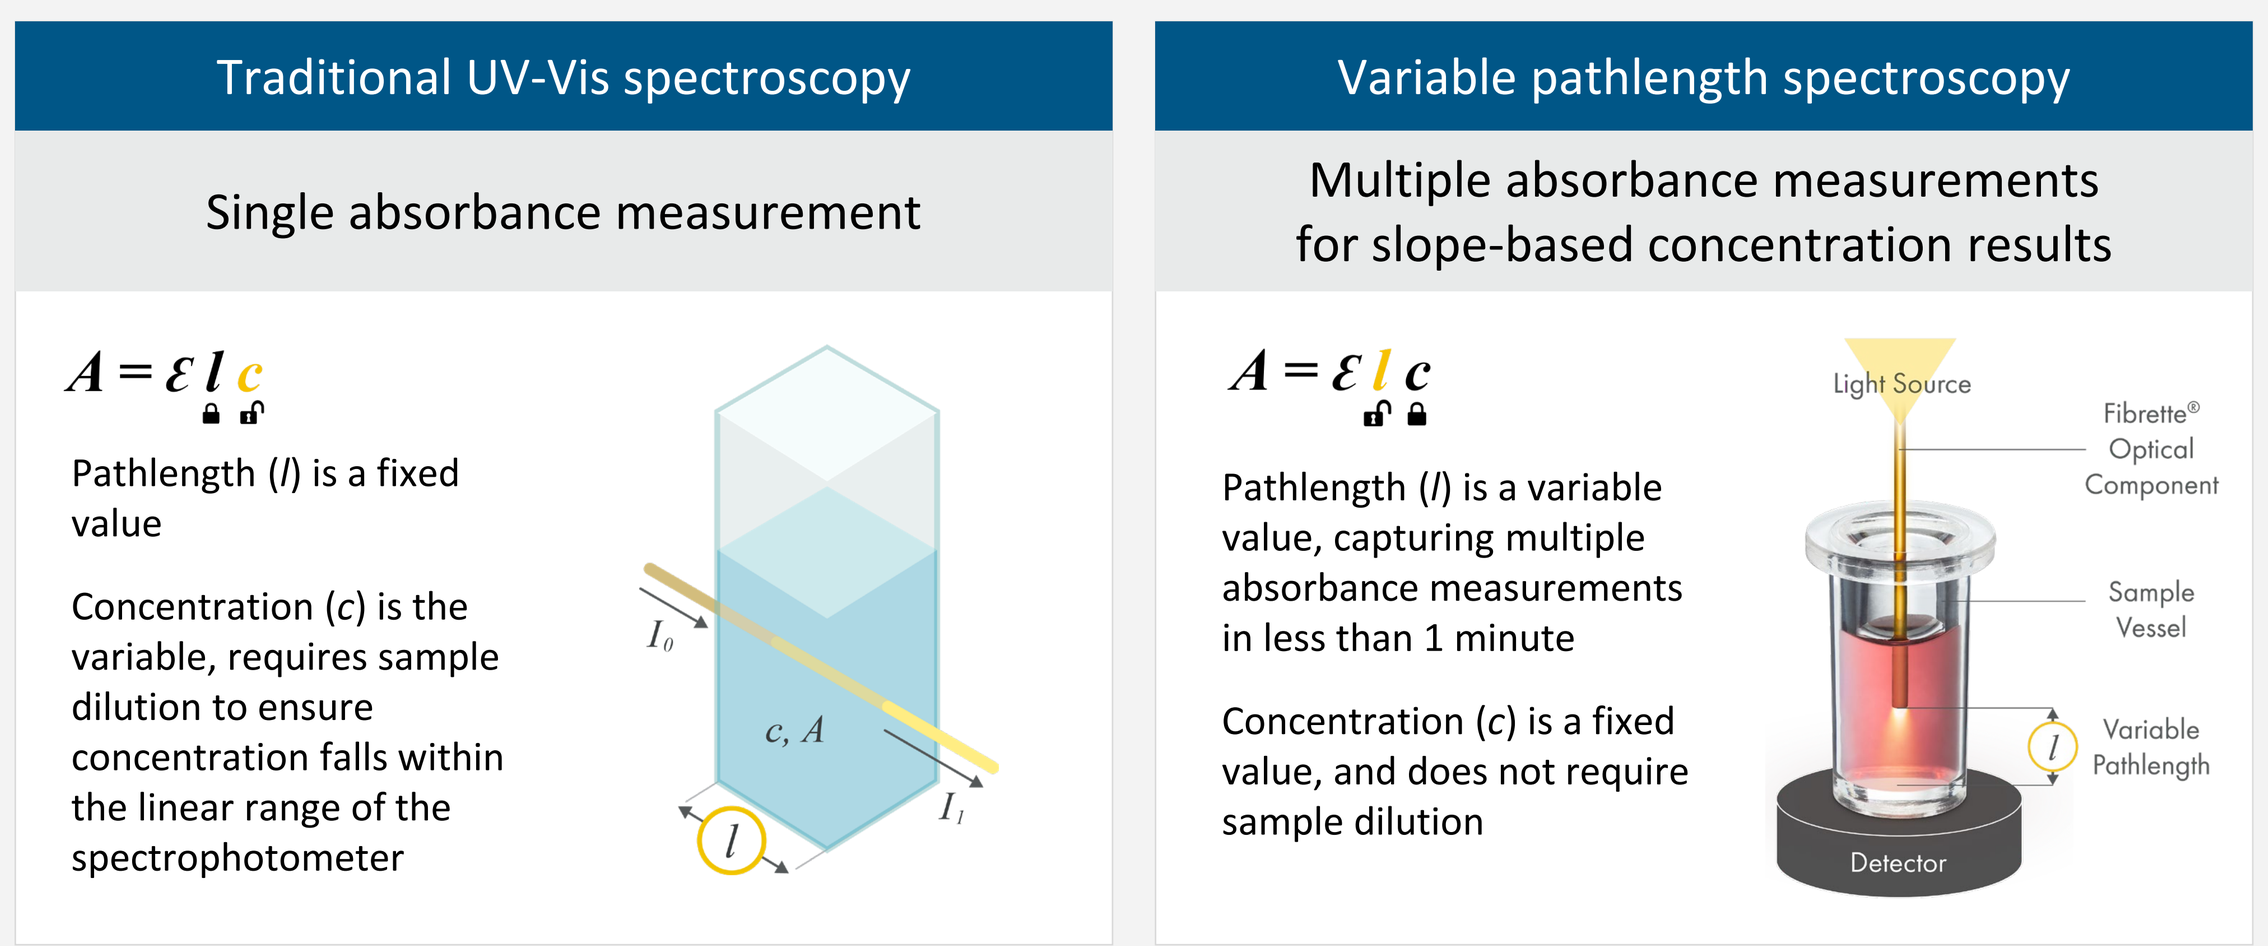
Traditional UV-Vis spectroscopy
Single absorbance measurement
A = ε l c
Pathlength (l) is a fixed value
Concentration (c) is the variable, requires sample dilution to ensure concentration falls within the linear range of the spectrophotometer
c, A
Variable pathlength spectroscopy
Multiple absorbance measurements for slope-based concentration results
A = ε l c
Pathlength (l) is a variable value, capturing multiple absorbance measurements in less than 1 minute
Concentration (c) is a fixed value, and does not require sample dilution
Light Source
Fibrette® Optical Component
Sample Vessel
Variable Pathlength
Detector

Beer's law

$A = \mathcal{E}\, l\, c$
$m = A/l$
$m = \mathcal{E}\, c$

A = absorbance
Ɛ = extinction coefficient
l = pathlength
c = concentration
m = slope

Figure 1. Comparison of fixed pathlength spectroscopy and variable pathlength spectroscopy with application of Beer–Lambert law.

$$m_{280}^{full} = m_{280}^{C} + m_{280}^{D}$$
$$m_{280}^{full} = m_{260}^{C} + m_{260}^{D}$$

$$m = \varepsilon \cdot c$$

$$m_{280}^{full} = \varepsilon_{280}^{C} \cdot conc^{C} + \varepsilon_{280}^{D} \cdot conc^{D}$$
$$m_{260}^{full} = \varepsilon_{260}^{C} \cdot conc^{C} + \varepsilon_{260}^{D} \cdot conc^{D}$$

$$conc^{D} = \frac{\varepsilon_{260}^{C} \cdot m_{280}^{full} - \varepsilon_{280}^{D} \cdot m_{260}^{full}}{\varepsilon_{260}^{C} \cdot \varepsilon_{280}^{D} - \varepsilon_{280}^{C} \cdot \varepsilon_{260}^{D}} = \frac{\varepsilon_{260}^{C} \cdot m_{280}^{full} - \varepsilon_{280}^{C} \cdot m_{260}^{full}}{\varepsilon_{260}^{C} \cdot \varepsilon_{280}^{D} - \varepsilon_{280}^{C} \cdot \varepsilon_{260}^{D}} \times \frac{1}{MW_{DNA}}$$

$$conc^{C} = \frac{\varepsilon_{260}^{D} \cdot m_{280}^{full} - \varepsilon_{280}^{D} \cdot m_{260}^{full}}{\varepsilon_{260}^{D} \cdot \varepsilon_{280}^{C} - \varepsilon_{280}^{D} \cdot \varepsilon_{260}^{C}} = \frac{\varepsilon_{260}^{D} \cdot m_{280}^{full} - \varepsilon_{280}^{D} \cdot m_{260}^{full}}{\varepsilon_{260}^{D} \cdot \varepsilon_{280}^{C} - \varepsilon_{280}^{D} \cdot \varepsilon_{260}^{C}} \times \frac{1}{MW_{C}}$$

$$\rightarrow$$

$$R = \frac{\varepsilon_{260}^{C}}{\varepsilon_{280}^{C}} \qquad K = \frac{\varepsilon_{280}^{D}}{\varepsilon_{260}^{D}}$$

$$\rightarrow$$

$$conc^{D} = \frac{R \cdot m_{280} - m_{260}}{\varepsilon_{260}^{D} \cdot (R \cdot K - 1)} \times N_A$$

$$conc^{C} = \frac{m_{280} - K \cdot m_{260}}{\varepsilon_{280}^{C} \cdot (1 - R \cdot K)} \times N_A$$

$$\downarrow$$

$$Empty/Full\ Ratio = \frac{conc^{C} - conc^{D}}{conc^{D}}$$

m^{full} = total slope absorbances, c= capsids, D = DNA, m^{c}= slope absorbance from capsid proteins, m^{D} = slope absorbance from DNA, ε = extinction coefficient, conc = concentration, MW = molecular weight,
R = ε_{260}^{C} / ε_{280}^{C} , K = ε_{280}^{D} / ε_{260}^{D} , N_A =Avogadro's constant.

Figure 2. AAV Titer Calculation based on slope data utilizing Variable Pathlength Technology at UV 260 nm and UV 280 nm wavelengths.

Accuracy

Full and empty AAV2, AAV8, and AAV9 standards were used to assess the accuracy of the SoloVPE System. The system compared the various serotypes against qPCR and ELISA to evaluate differences in viral genome titer as well as viral capsid titer.

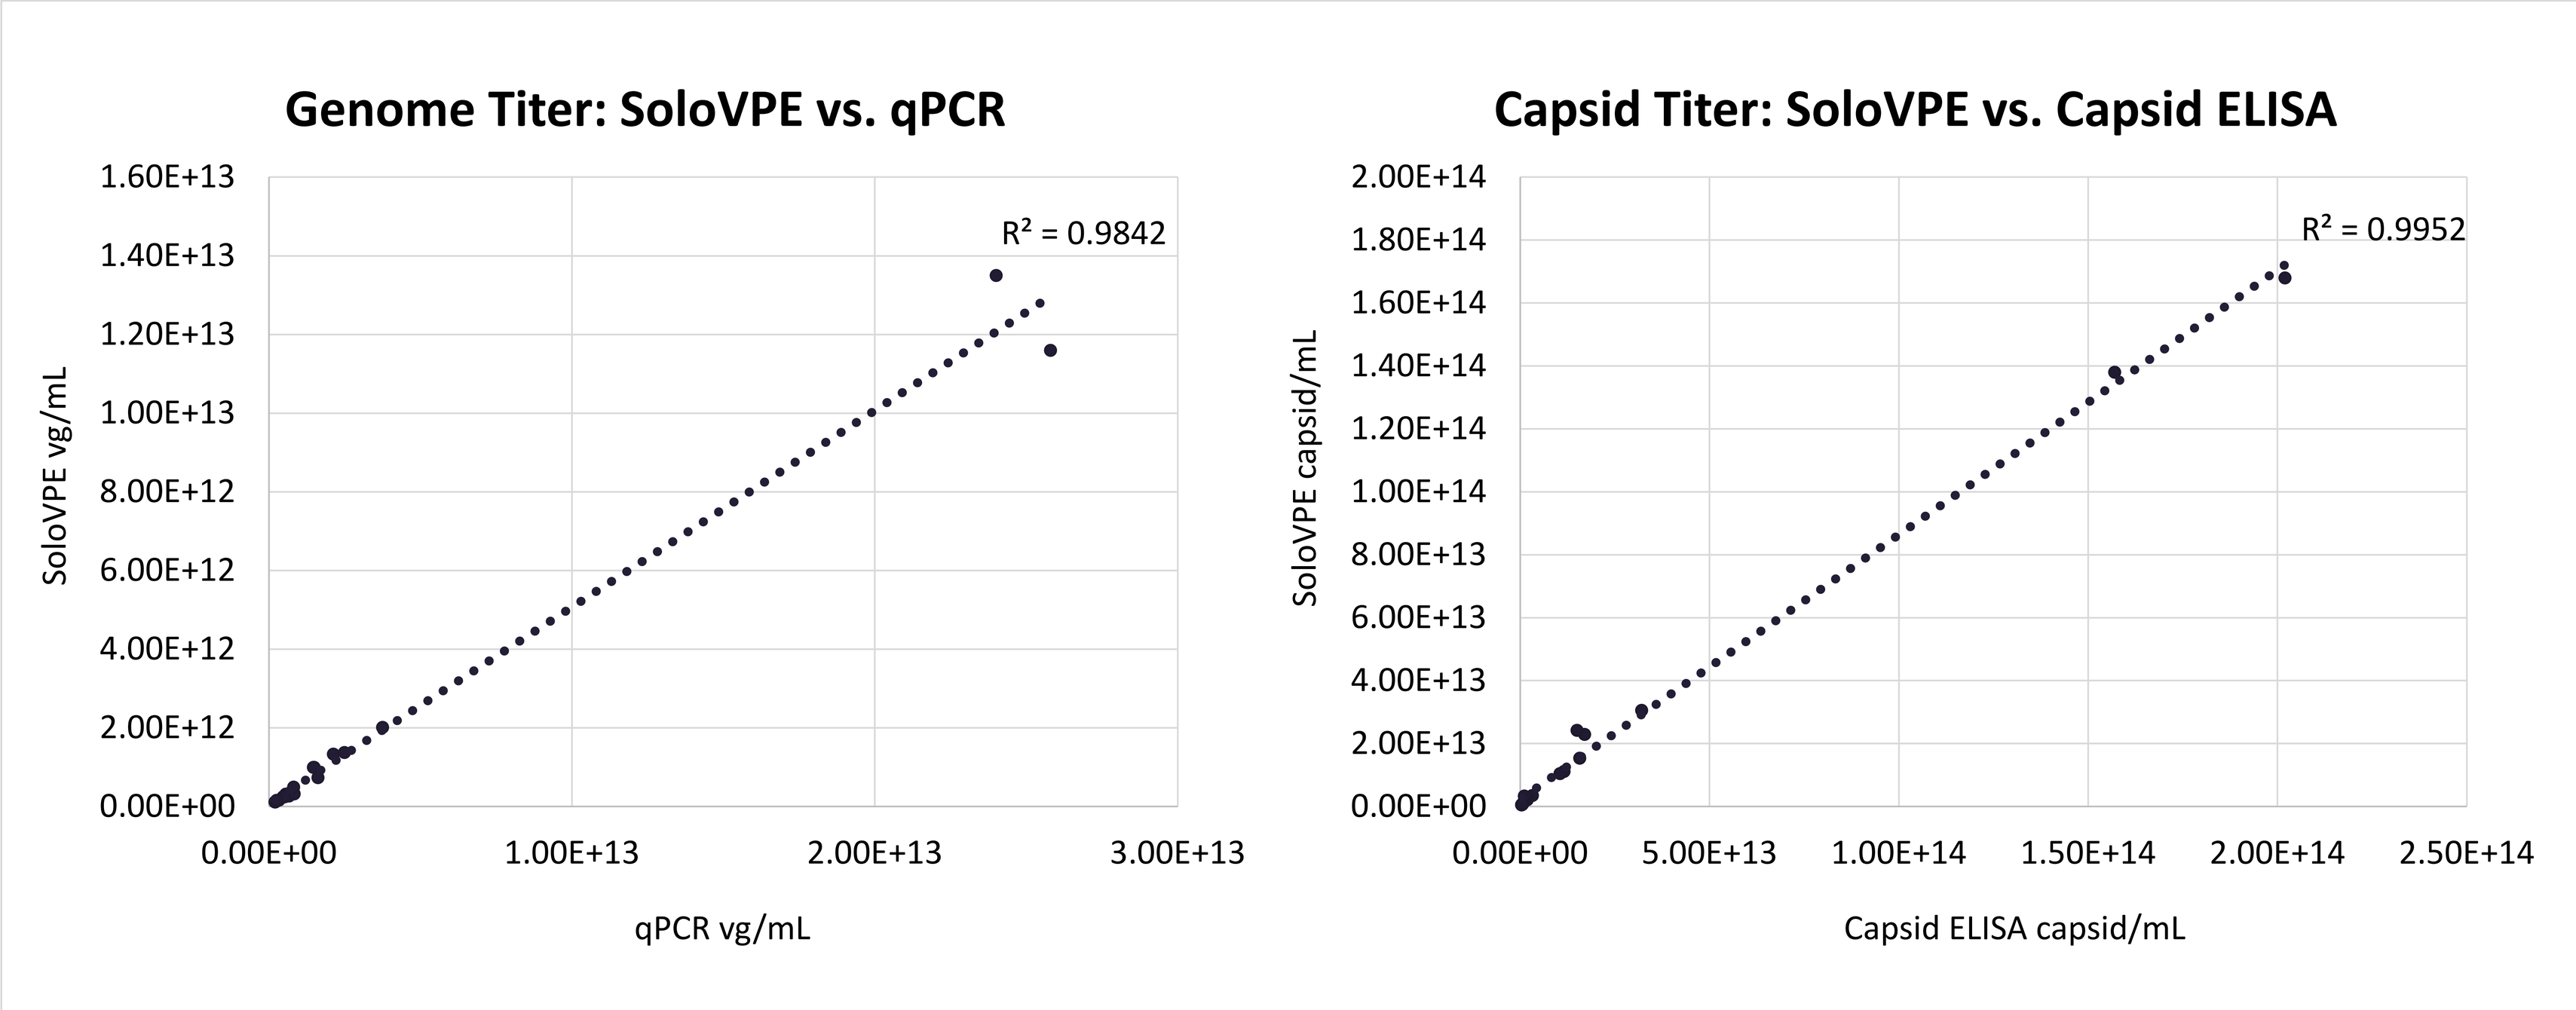
Genome Titer: SoloVPE vs. qPCR
R² = 0.9842
SoloVPE vg/mL
qPCR vg/mL
1.60E+13
1.40E+13
1.20E+13
1.00E+13
8.00E+12
6.00E+12
4.00E+12
2.00E+12
0.00E+00
1.00E+13
2.00E+13
3.00E+13
Capsid Titer: SoloVPE vs. Capsid ELISA
R² = 0.9952
SoloVPE capsid/mL
Capsid ELISA capsid/mL
2.00E+14
1.80E+14
1.60E+14
1.40E+14
1.20E+14
1.00E+14
8.00E+13
6.00E+13
4.00E+13
2.00E+13
0.00E+00
5.00E+13
1.00E+14
1.50E+14
2.00E+14
2.50E+14

Figure 3. AAV titer for downstream intermediate samples calculated using VPT shows excellent agreement with qPCR and Capsid ELISA data in a linear trend

Linearity, Repeatability, and Intermediate Precision

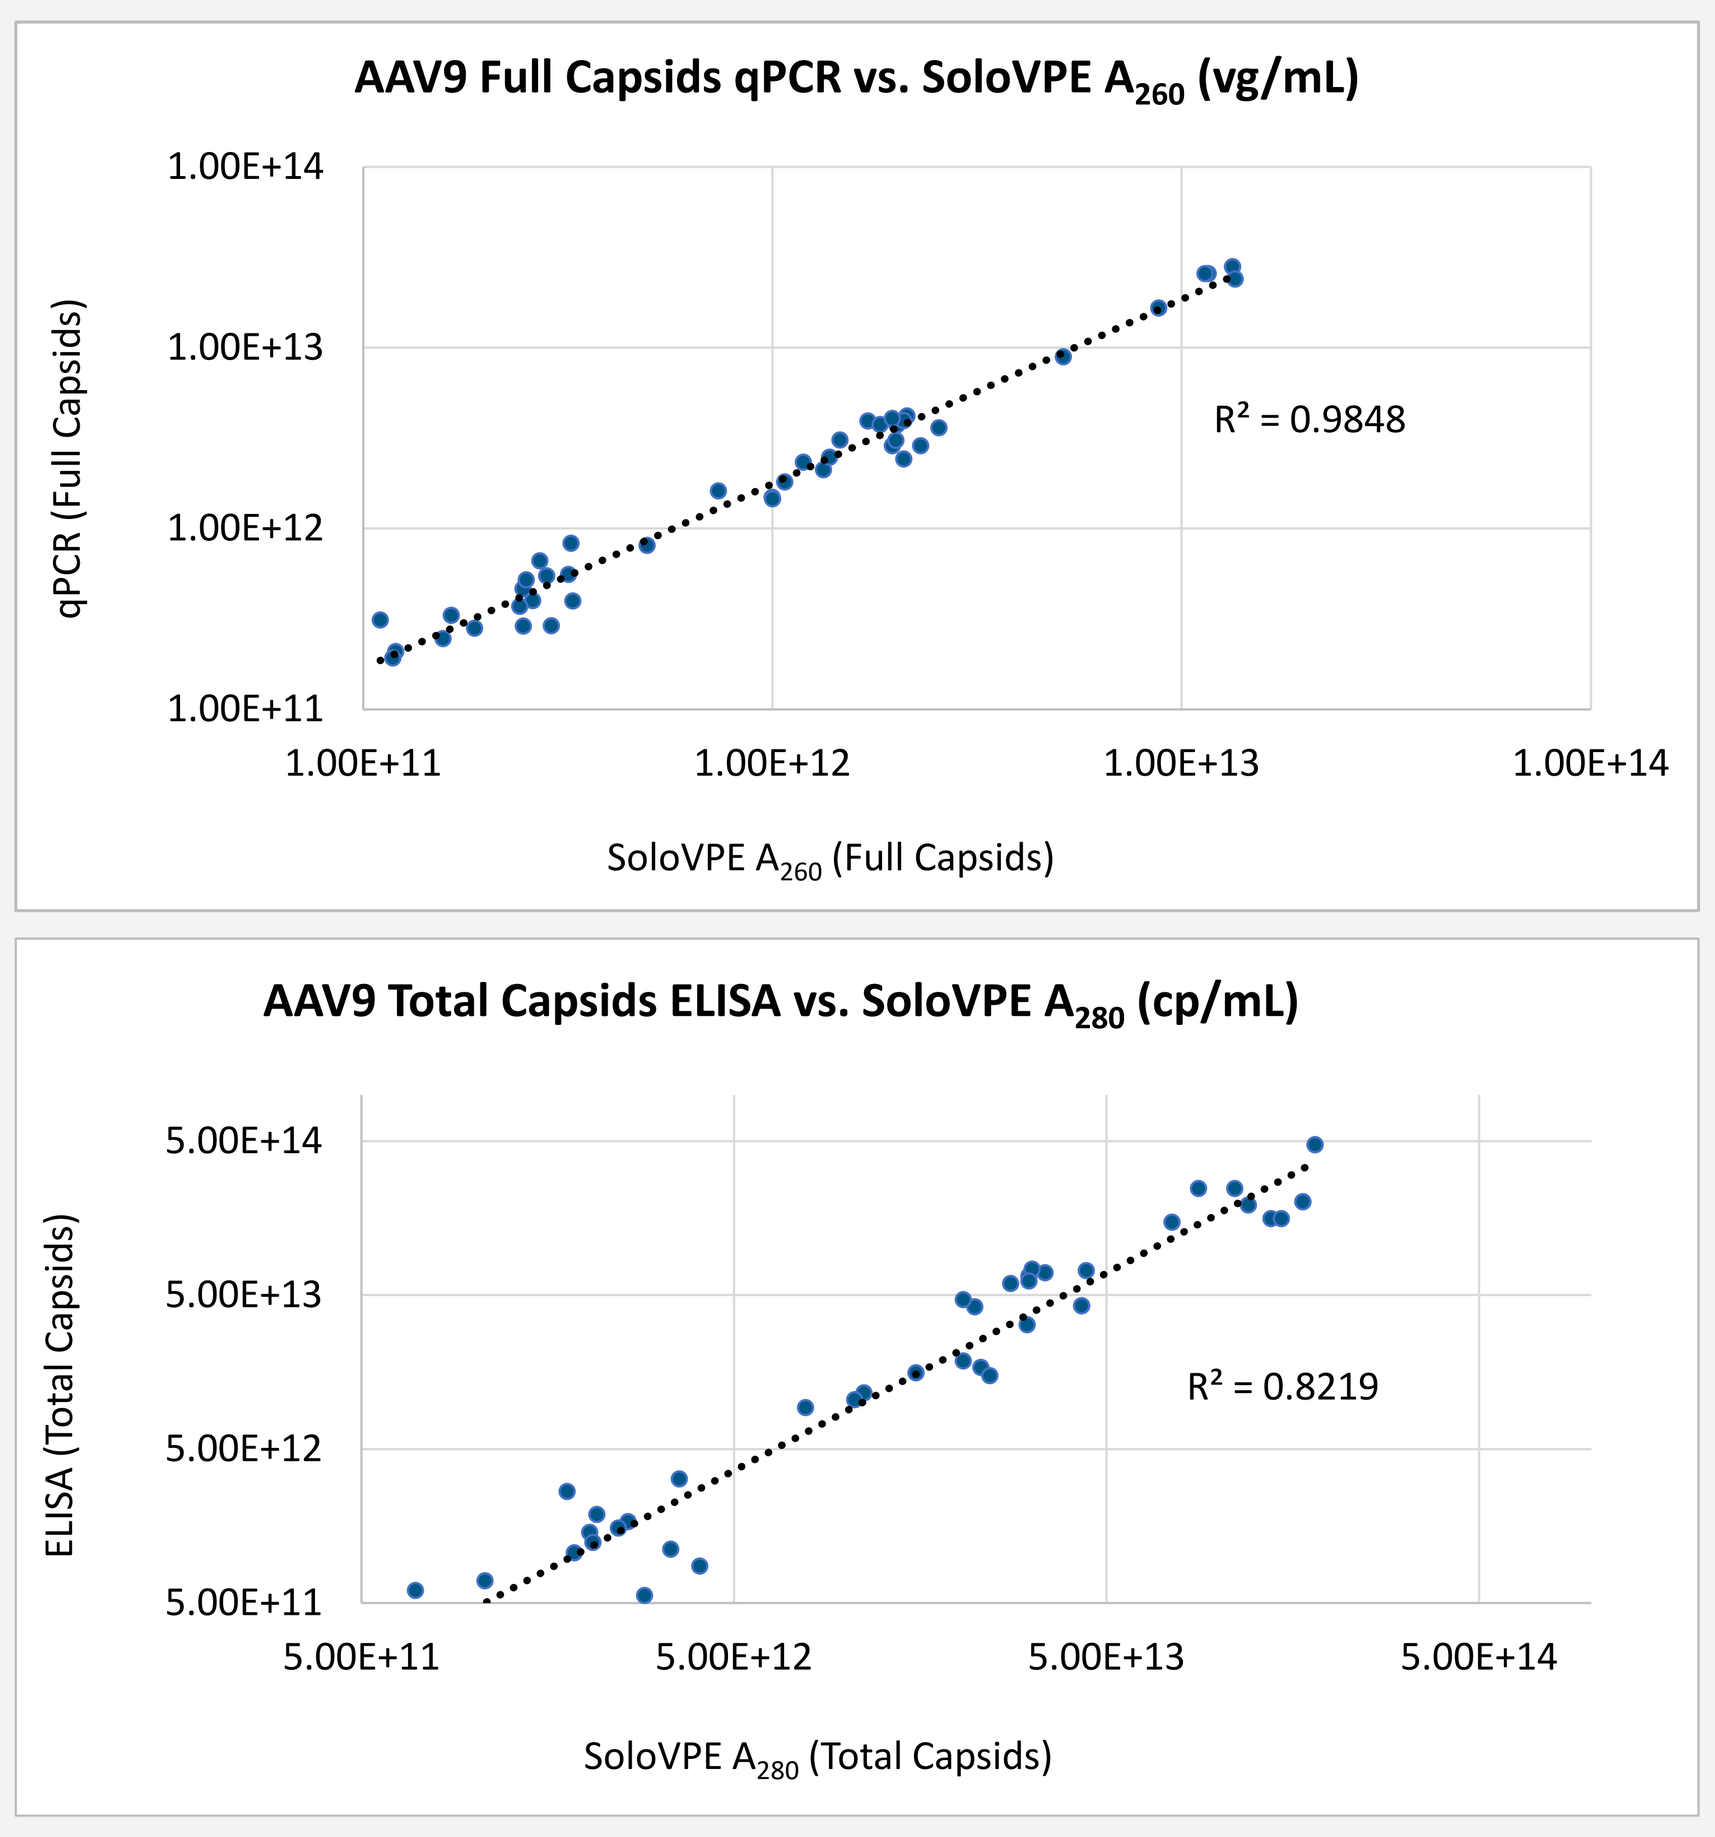
AAV9 Full Capsids qPCR vs. SoloVPE A260 (vg/mL)
qPCR (Full Capsids)
SoloVPE A260 (Full Capsids)
1.00E+14
1.00E+13
1.00E+12
1.00E+11
R² = 0.9848
AAV9 Total Capsids ELISA vs. SoloVPE A280 (cp/mL)
ELISA (Total Capsids)
SoloVPE A280 (Total Capsids)
5.00E+14
5.00E+13
5.00E+12
5.00E+11
R² = 0.8219

Table 1. Repeatability: The AAV samples were measured in triplicates, using the same Fibrette and the same aliquoted sample within the same sample vessel.

Sample	Sample Repeats	m260	Mean m260	RSD	m280	Mean m280	RSD
1	R-1	0.28146	0.28101	0.15%	0.26049	0.26021	0.19%
	R-2	0.28094			0.26048		
	R-3	0.28064			0.25965		
2	R-1	0.09246	0.09247	0.03%	0.13438	0.13456	0.18%
	R-2	0.09250			0.13483		
	R-3	0.09245			0.13448		

Table 2. Intermediate Precision: The AAV sample was measured by two scientists on different days using different Fibrettes, sample vessels, and aliquots.

Sample	m260	Mean m260	RSD	m280	Mean m280	RSD
N-1	0.28288	0.28183	0.52%	0.26820	0.26662	0.84%
N-2	0.28079			0.26504		

Figure 4. Comparison of variable pathlength spectroscopy to qPCR and ELISA. Concentration range tested: qPCR: 4.7 E+11 – 2.6 E+13 vg/mL and Capsid ELISA: 2.9 E+12 – 1.6 E+14 capsid/mL.

VPT in Downstream Process Development Applications: Chromatography Steps and Ultrafiltration/Diafiltration (UF/DF)

We assessed the viability of VPT for rAAV vector and capsid titer measurement as well as empty/full capsid ratio determination during downstream process development. VPT has demonstrated its value as a process analytical technology (PAT) tool to provide real-time feedback to ensure step recovery and to meet target titer requirement in the UF/DF step.

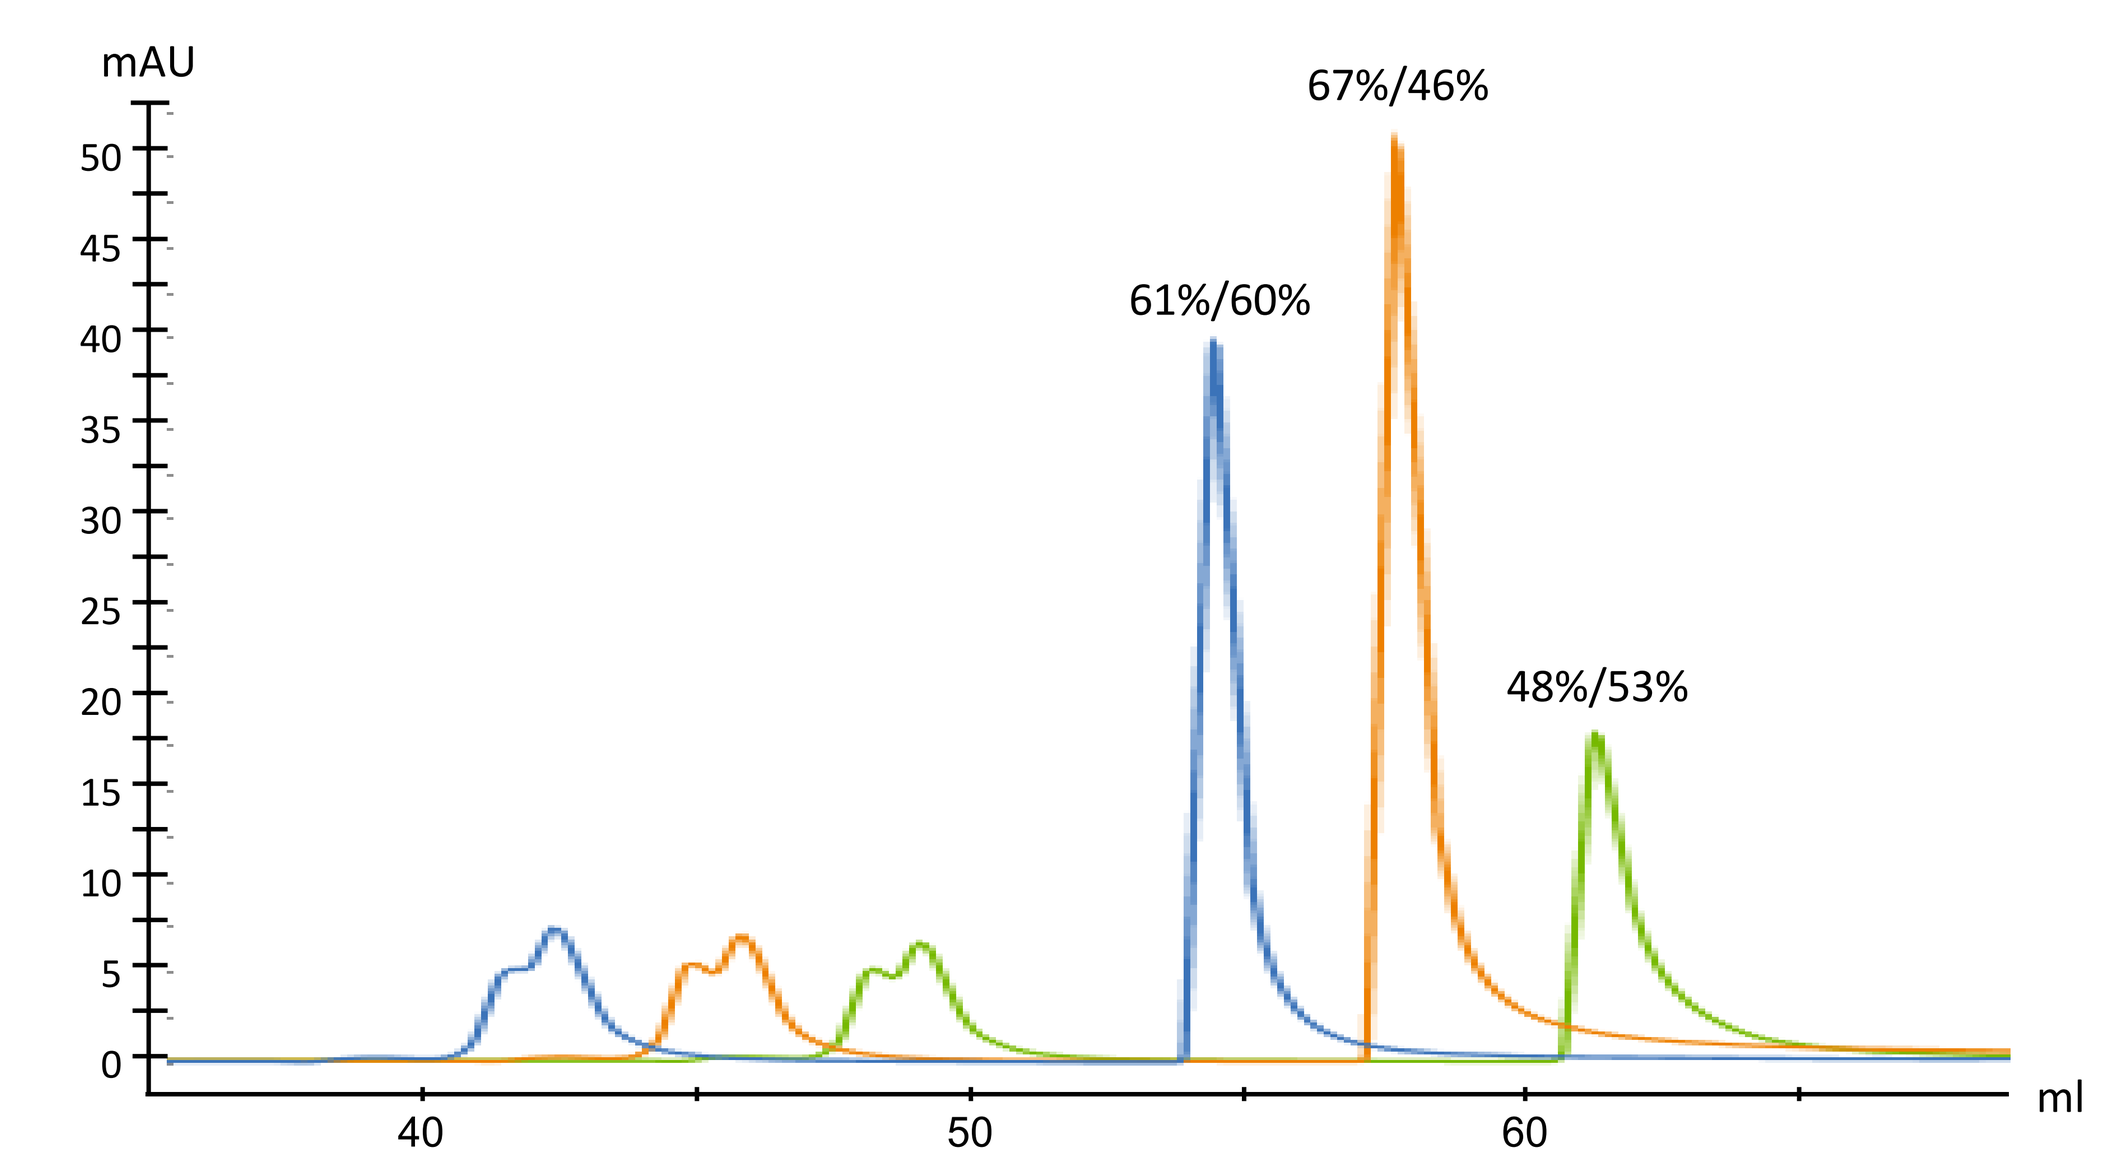
mAU
50
45
40
35
30
25
20
15
10
5
0
40
50
60
ml
67%/46%
61%/60%
48%/53%

Potential Applications
Load pH/conductivity
Residence time
Resin screening
Loading capacity
Buffer additives
Elution buffer pH conductivity
Gradient/step elution

Figure 5. Elution Buffer Conductivity Screening– Recovery/Full%. Overlay of elution peaks from different screening runs demonstrated VPT measurement of vector titer and full capsid % allows for quick decision making in AAV polishing step process development based on recovery and full capsid enrichment. Rapid analytics provided with the SoloVPE System may be apply to other downstream applications.

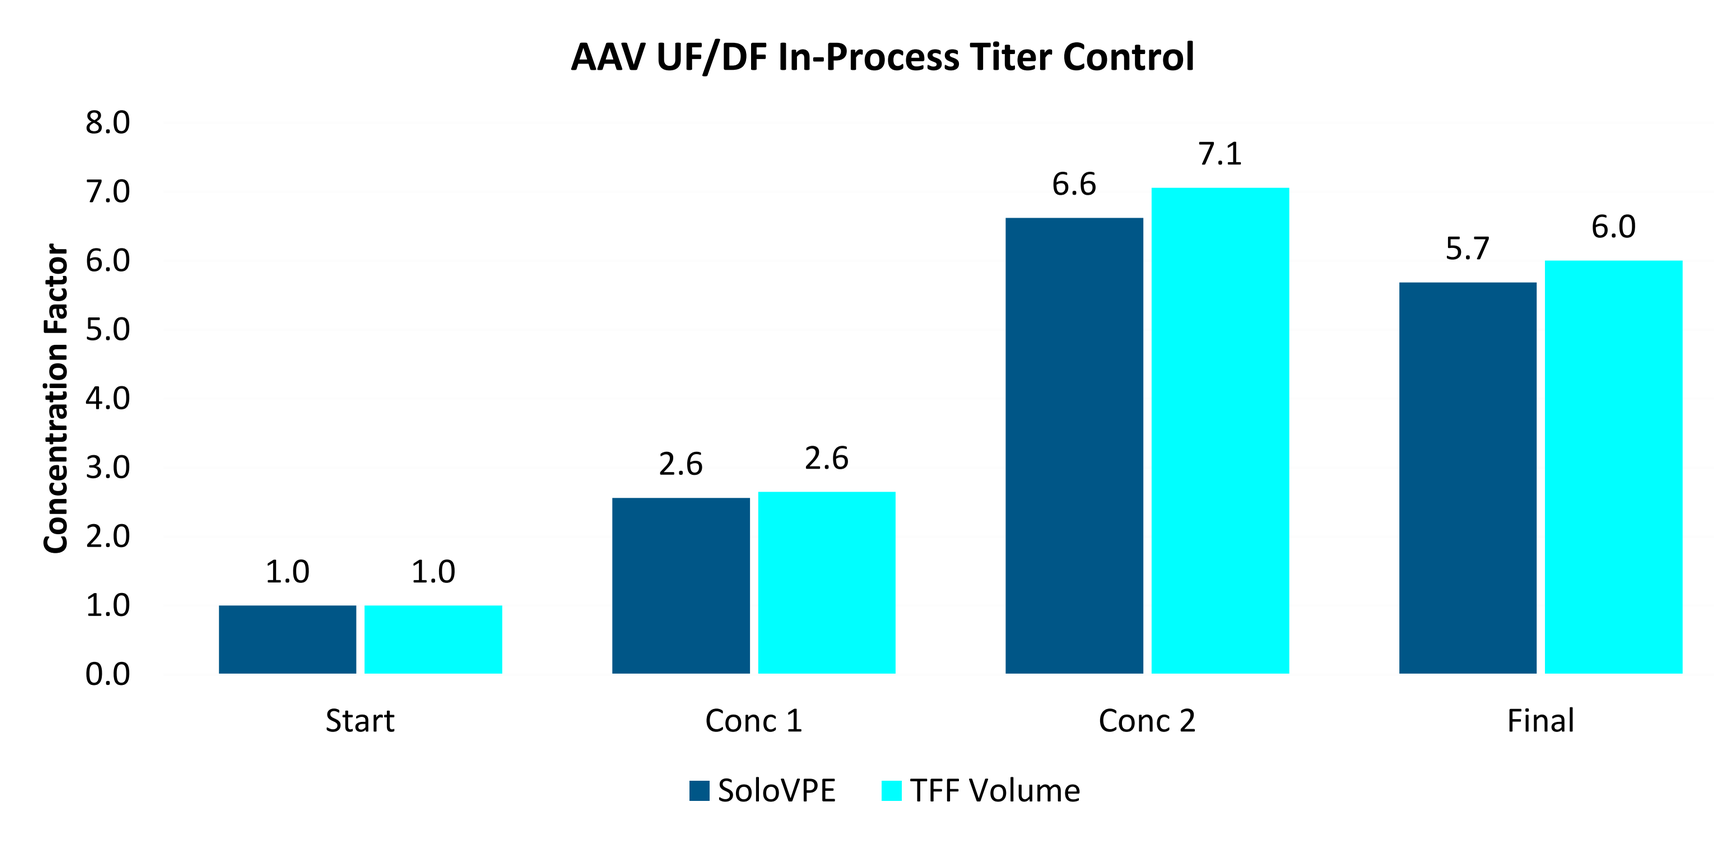
AAV UF/DF In-Process Titer Control
Concentration Factor
8.0
7.0
6.0
5.0
4.0
3.0
2.0
1.0
0.0
1.0
1.0
2.6
2.6
6.6
7.1
5.7
6.0
Start
Conc 1
Conc 2
Final
SoloVPE
TFF Volume

Figure 6. VPT application in UF/DF step. The concentration factor obtained through the VPT method aligns well with the calculation based on volume reduction during TFF run using the KrosFlo® KR2i TFF System. This finding suggests that the SoloVPE titer assay can be used as a practical method for process control during the UF/DF step.

Conclusion

The development of recombinant adeno-associated virus (rAAV) processes necessitates the availability of a rapid and dependable method for measuring vector and capsid titers. The application of Variable Pathlength Technology (VPT) for titer determination has yielded comparable results to those obtained through qPCR titer and Capsid ELISA titer analyses performed on purified samples. A comprehensive evaluation of VPT has demonstrated its commendable accuracy, linearity, repeatability, and intermediate precision. In comparison to the prevailing technologies currently employed in the industry, VPT offers a substantial advantage in terms of time to results and user friendliness. A triplicate-combined measurement of viral genome and capsid titers can be completed in less than five minutes, rendering this method highly suitable as an at-line test during downstream process development, enabling real-time decision-making.

References

1. Sommer, J., et al. (2003). Quantification of Adeno-Associated Virus Particles and Empty Capsids by Optical Density Measurement. Molecular Therapy, 7, 122-128.

2. Harald, E., et al. (2022). Using Slope Spectroscopy to Streamline Development for AAV Titer Determination Processes. American Pharmaceutical Review, April 18, 2022.

Learn more at **repligen.com**

© 2023 Repligen Corporation. All rights reserved. The trademarks mentioned herein are the property of Repligen Corporation and/or its affiliate(s) or their respective owners. | DOC0330 eRev. 1.0 11/13/2023